Magnetic relaxometry detection of stealth, antibody-targeted micellar iron oxide nanoparticles *in vivo*

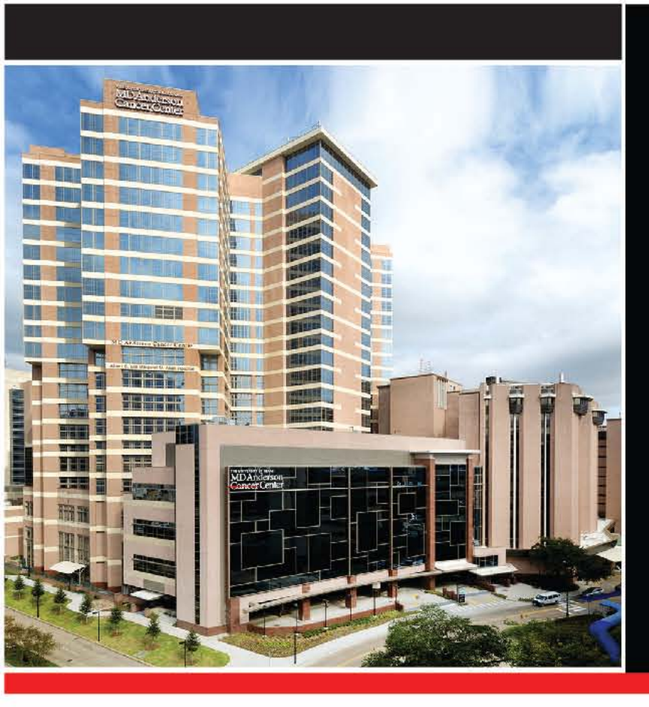

Rebeca Romero Aburto[1], Konstantin Sokolov[1], Adam M. Kulp[1], Erika C.Vreeland[3], Zhen Lu[2], Robert C. Bast, Jr[2], John D. Hazle[1], and Kelsey B. Mathieu[1].
1 Department of Imaging Physics, MD Anderson Cancer Center, Houston, TX, USA
2 Department of Experimental Therapeutics, MD Anderson Cancer Center, Houston, TX, USA
3 Imagion Biosystems, Inc. Albuquerque, NM, USA

Introduction

By sensing changes in Neél relaxation of iron oxide (Fe_3O_4) particles targeted to cancer biomarkers, magnetic relaxometry (MRX), has the potential to provide unprecedented sensitivity in cancer detection[1,2]. To achieve this, a stealth, monodisperse nanoformulation with a long circulation persistence is required. Our strategy consist in developing magnetic micelles via direct infusion of an organic phase mixture of phospholipids and Fe_3O_4 nanoparticles into water (**Figure 1A,B**).

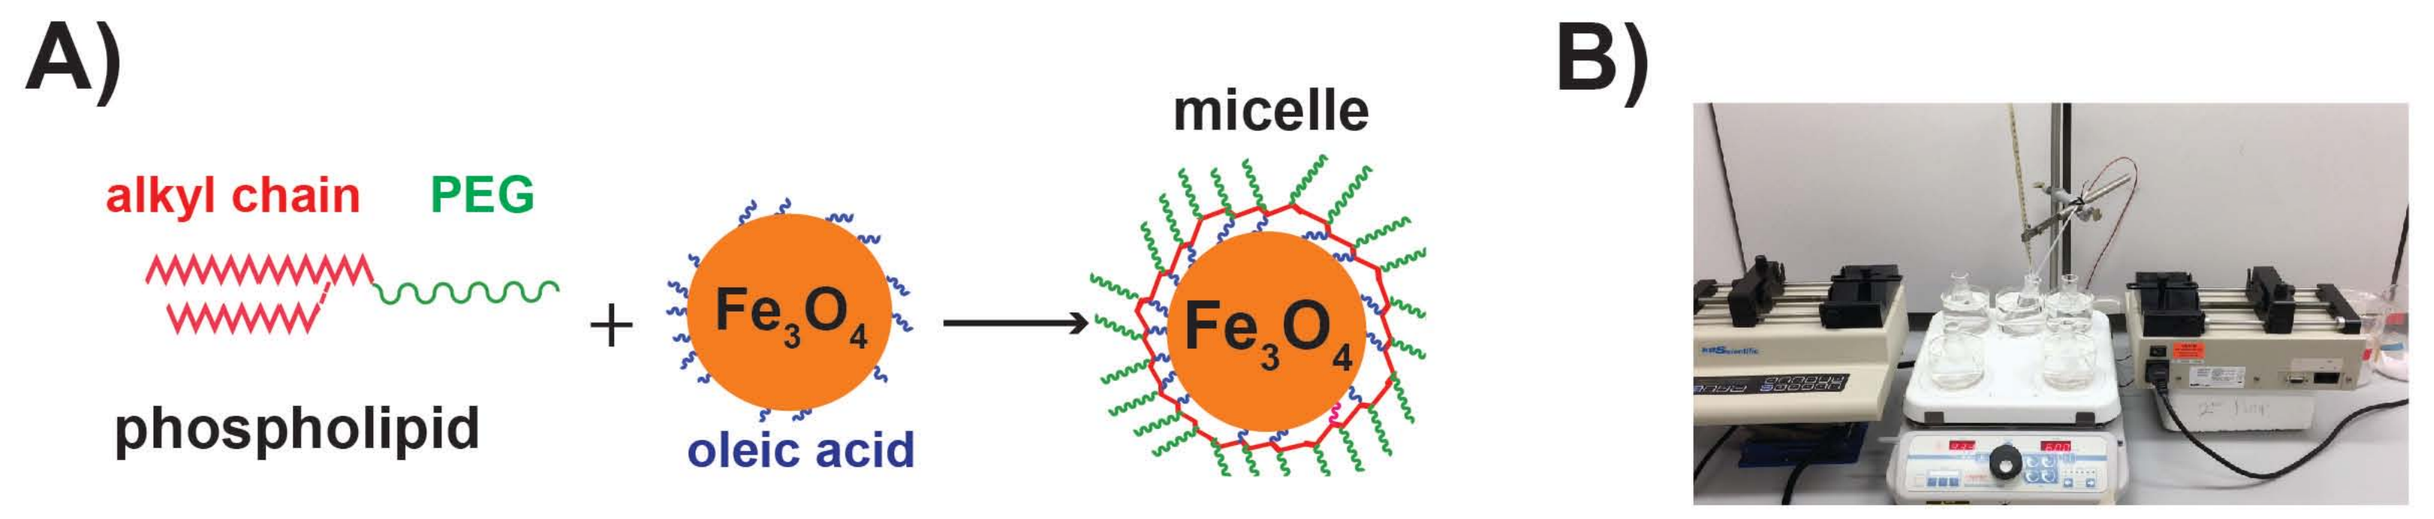
A)
alkyl chain
PEG
phospholipid
oleic acid
micelle
B)

Figure 1. Surface modification to yield hydrophilic Fe_3O_4 micelles

Results and Discussion

Hydrophobic interactions between the phospholipid alkyl chain and the oleic acid on the surface of Fe_3O_4 produce monodisperse micelles with a mean diameter of 75±12 nm, and a zeta potential of -10mV (**Figure 2A, B**). Direct infusion proved to be a reproducible technique, maximizing product yield (**Figure 2C**). Fe_3O_4 micelles were determined stable in various biological media, including human plasma, for at least 24 hours with no detectable protein adsorption (**Figure 2D**). Possibly indicating stealthiness against opsonization and prolonged *in vivo* circulation.

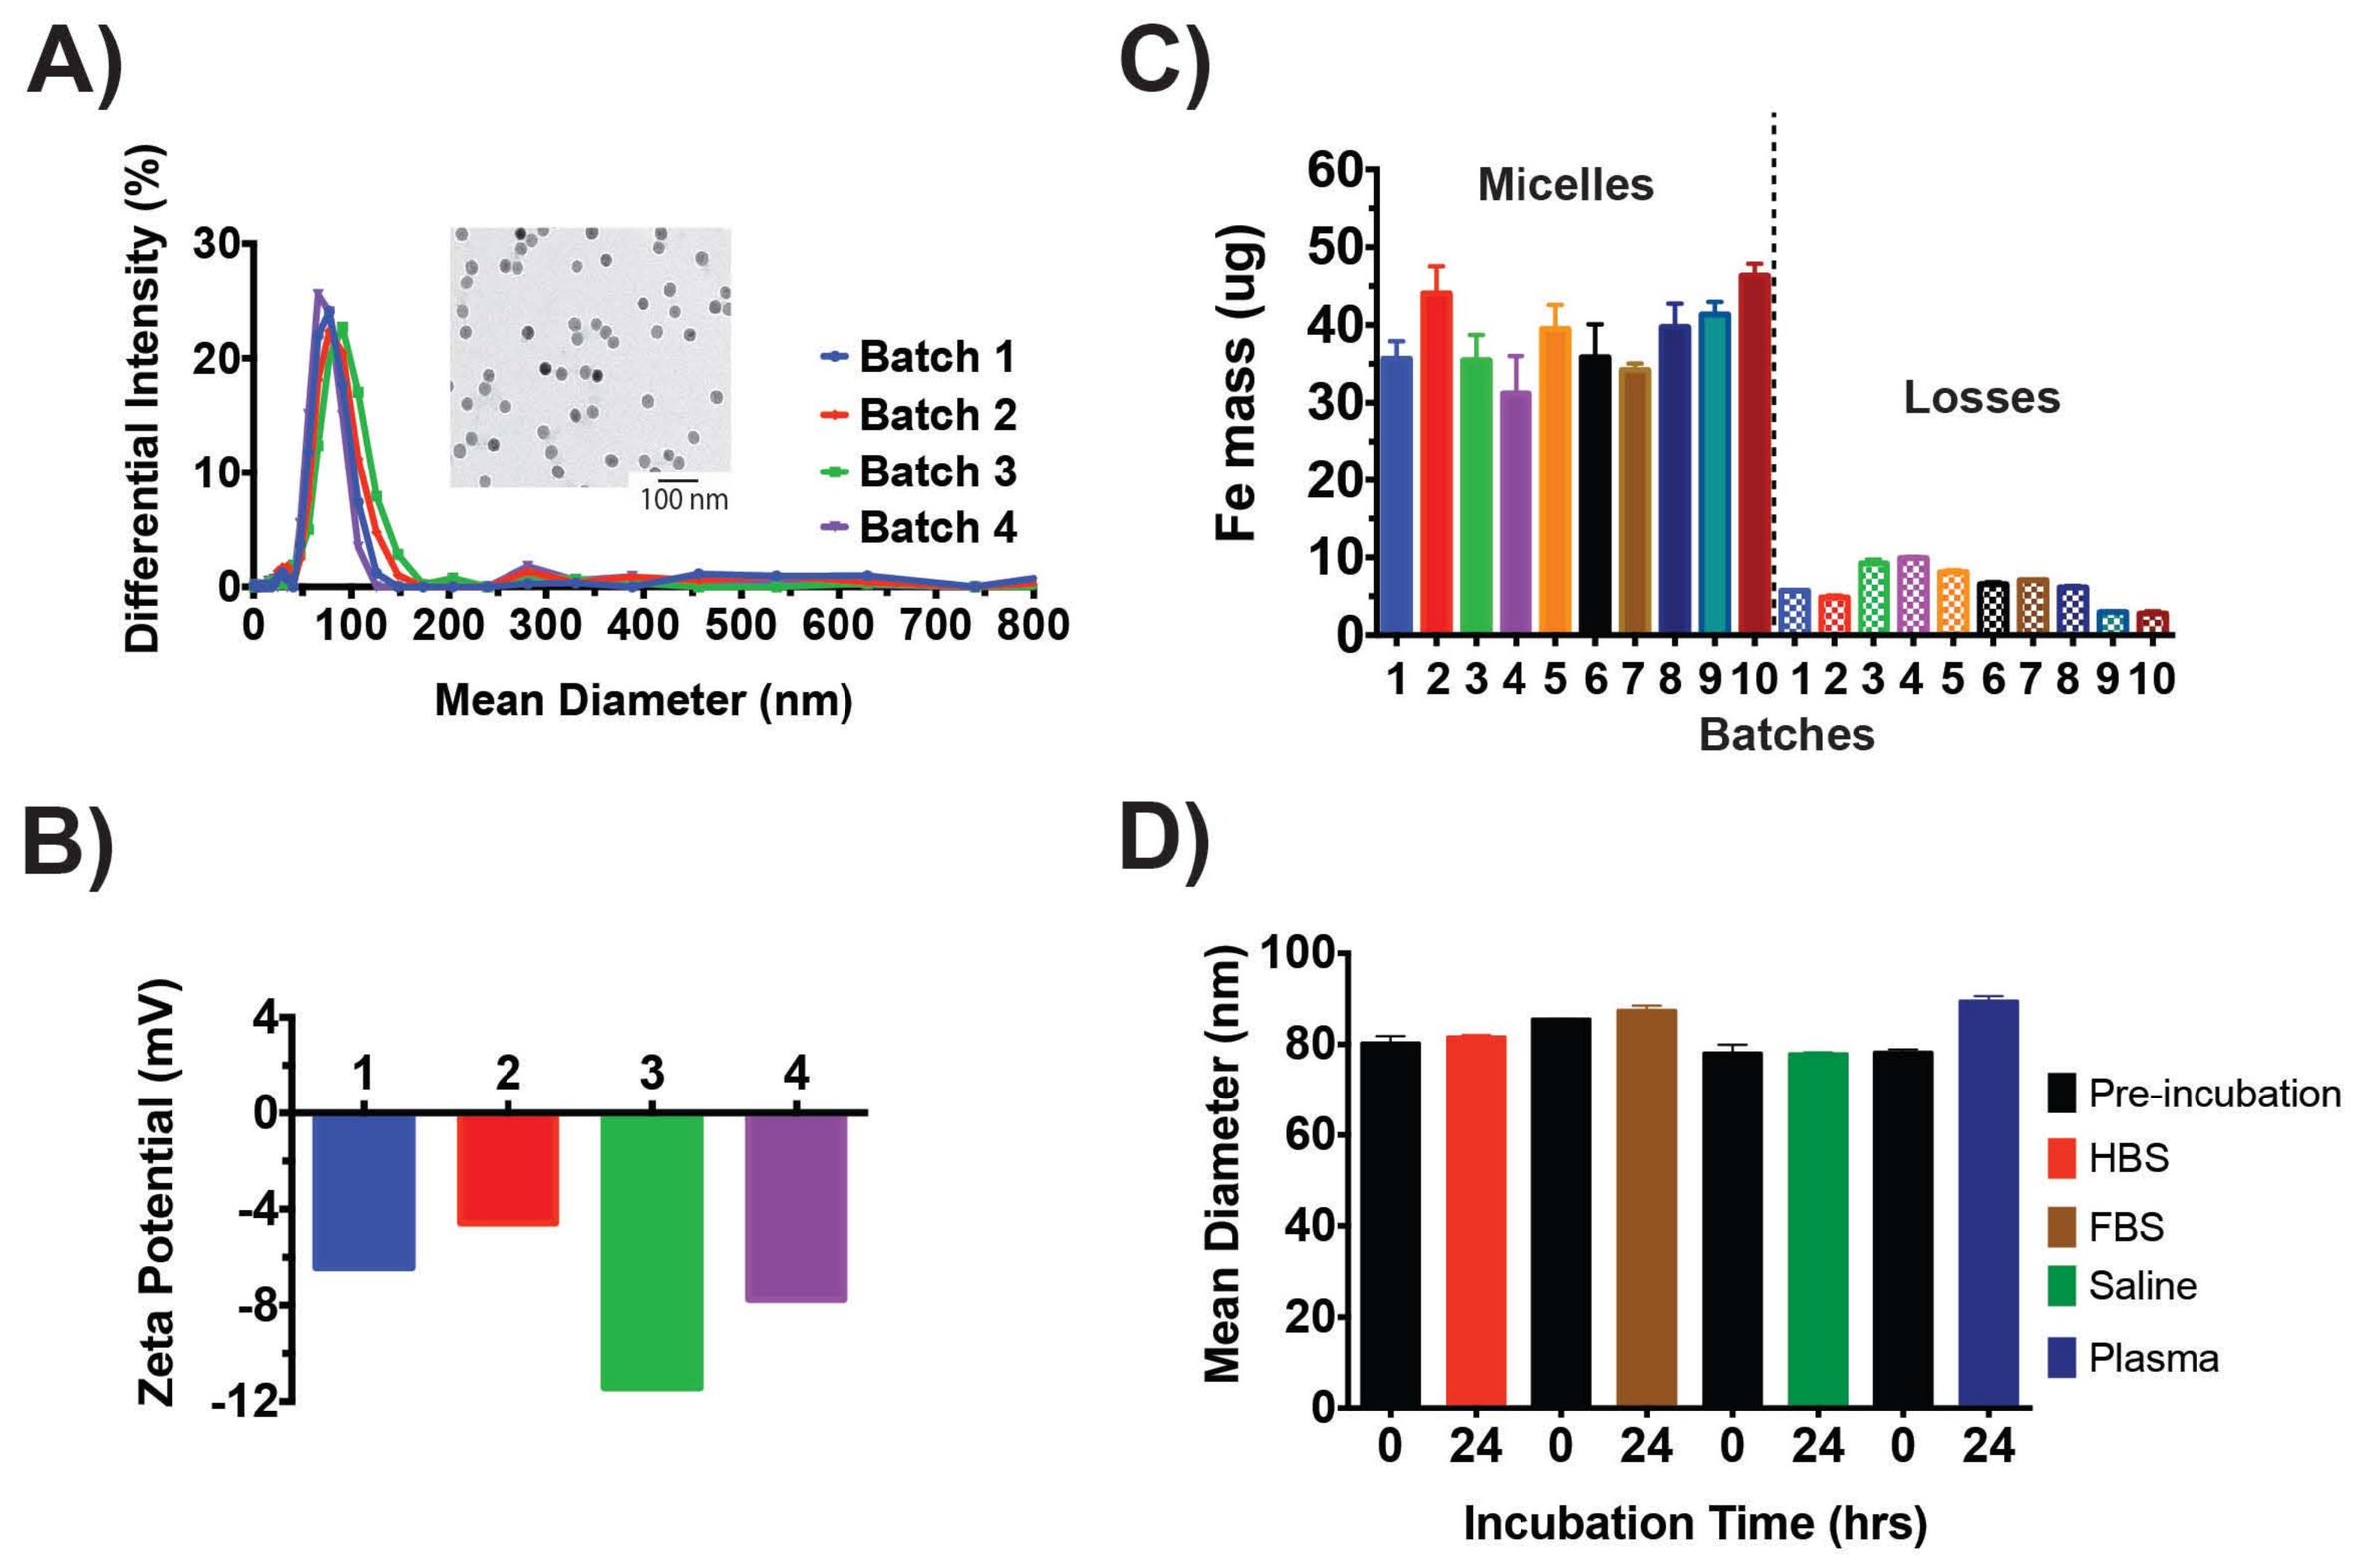
A)
Differential Intensity (%)
Mean Diameter (nm)
Batch 1
Batch 2
Batch 3
Batch 4
100 nm
B)
Zeta Potential (mV)
C)
Fe mass (ug)
Micelles
Losses
Batches
D)
Mean Diameter (nm)
Incubation Time (hrs)
Pre-incubation
HBS
FBS
Saline
Plasma

Figure 2. Micellar physico-chemical characterization
Size (**A**) and zeta potential (**B**) characterization via dynamic light scattering (DLS). Induced coupled plasma mass spectroscopy (ICP-MS) to determine iron mass per batch (**C**).Stability test after 24 hrs incubation of micelles using DLS (**D**).

To corroborate the stealthiness of the micellar formulation towards opsonization, iron uptaken by mouse macrophages was quantified via ICP-MS after 1 hr incubation at 37ºC (**Figure 3A**). Resulting in ~1% uptake, amounting to 0.2 μg of iron, these results support our initial observation. MRX detection was within the noise range (**Figure 3B**). *In vivo* circulation persistence study showed minimal MRX signal 2 hrs after intravenous injection into healthy mice, indicating that the micelles remained in circulation (**Figure 3D**). Serial blood draws were used to quantify iron in plasma to validate MRX results (**Figure 3C**).

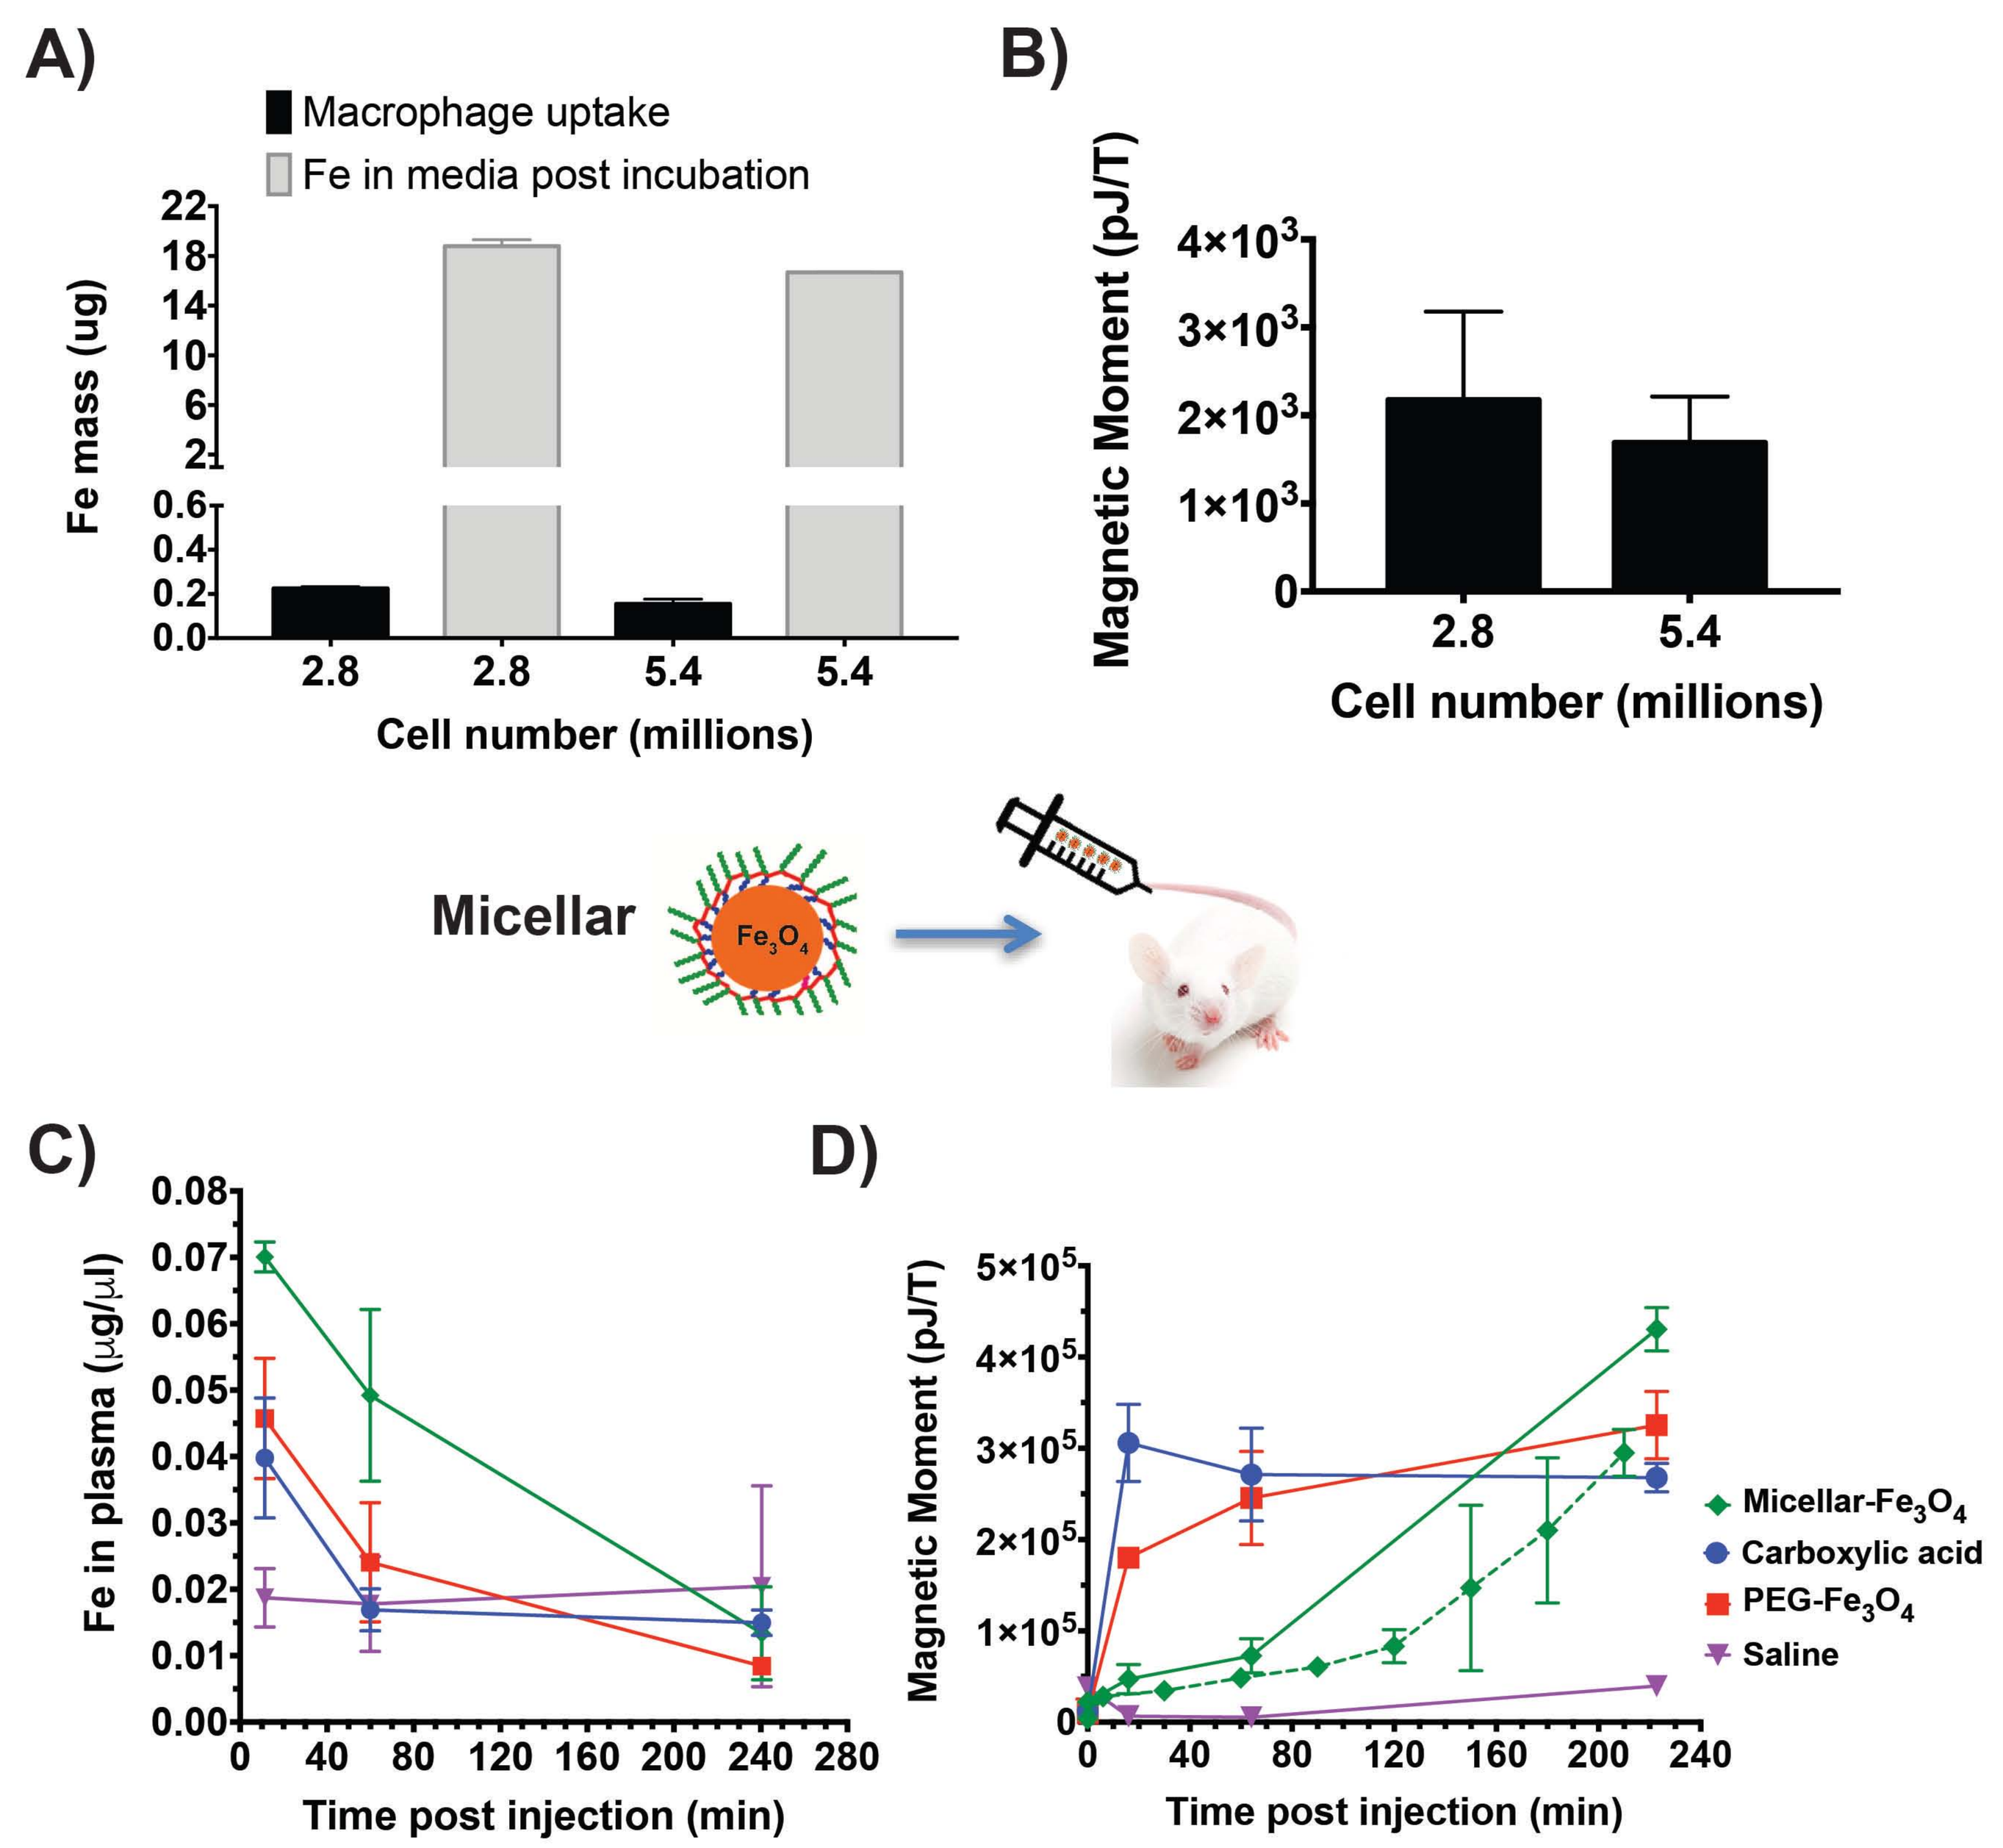
A)
Macrophage uptake
Fe in media post incubation
Fe mass (ug)
Cell number (millions)
B)
Magnetic Moment (pJ/T)
Cell number (millions)
Micellar
C)
Fe in plasma (μg/μl)
Time post injection (min)
D)
Magnetic Moment (pJ/T)
Time post injection (min)
Micellar-Fe3O4
Carboxylic acid
PEG-Fe3O4
Saline

Figure 3. Stealthiness and circulation persistence of micellar iron oxide
Mass of iron uptaken by macrophages after 1hr incubation in suspension (**A**). MRX signal of macrophage cell pellets (**B**). Iron concentration, determined by ICP-MS, in mouse plasma to survey circulation persistence (**C**). Mice scanned as a function of time under MRX (n=3) (**D**).

Specific cancer detection is only possible through targeted agents. Functional micelles can be produced with direct infusion, facilitating bio-conjugations. Directional conjugation maximizes antibody functionality and can be achieved by the following chemical modifications[3]. First, the hydroxyl group of the carbohydrate, ideally located in the Fc-portion of the antibody, will be oxidized to aldehyde (**Figure 4A**). Followed by the formation of a hydrazone bond between the linker and the antibody (**Figure 4B**). Finally, the antibody is linked to the micelle through a maleimide-thiol bond (**Figure 4C**).

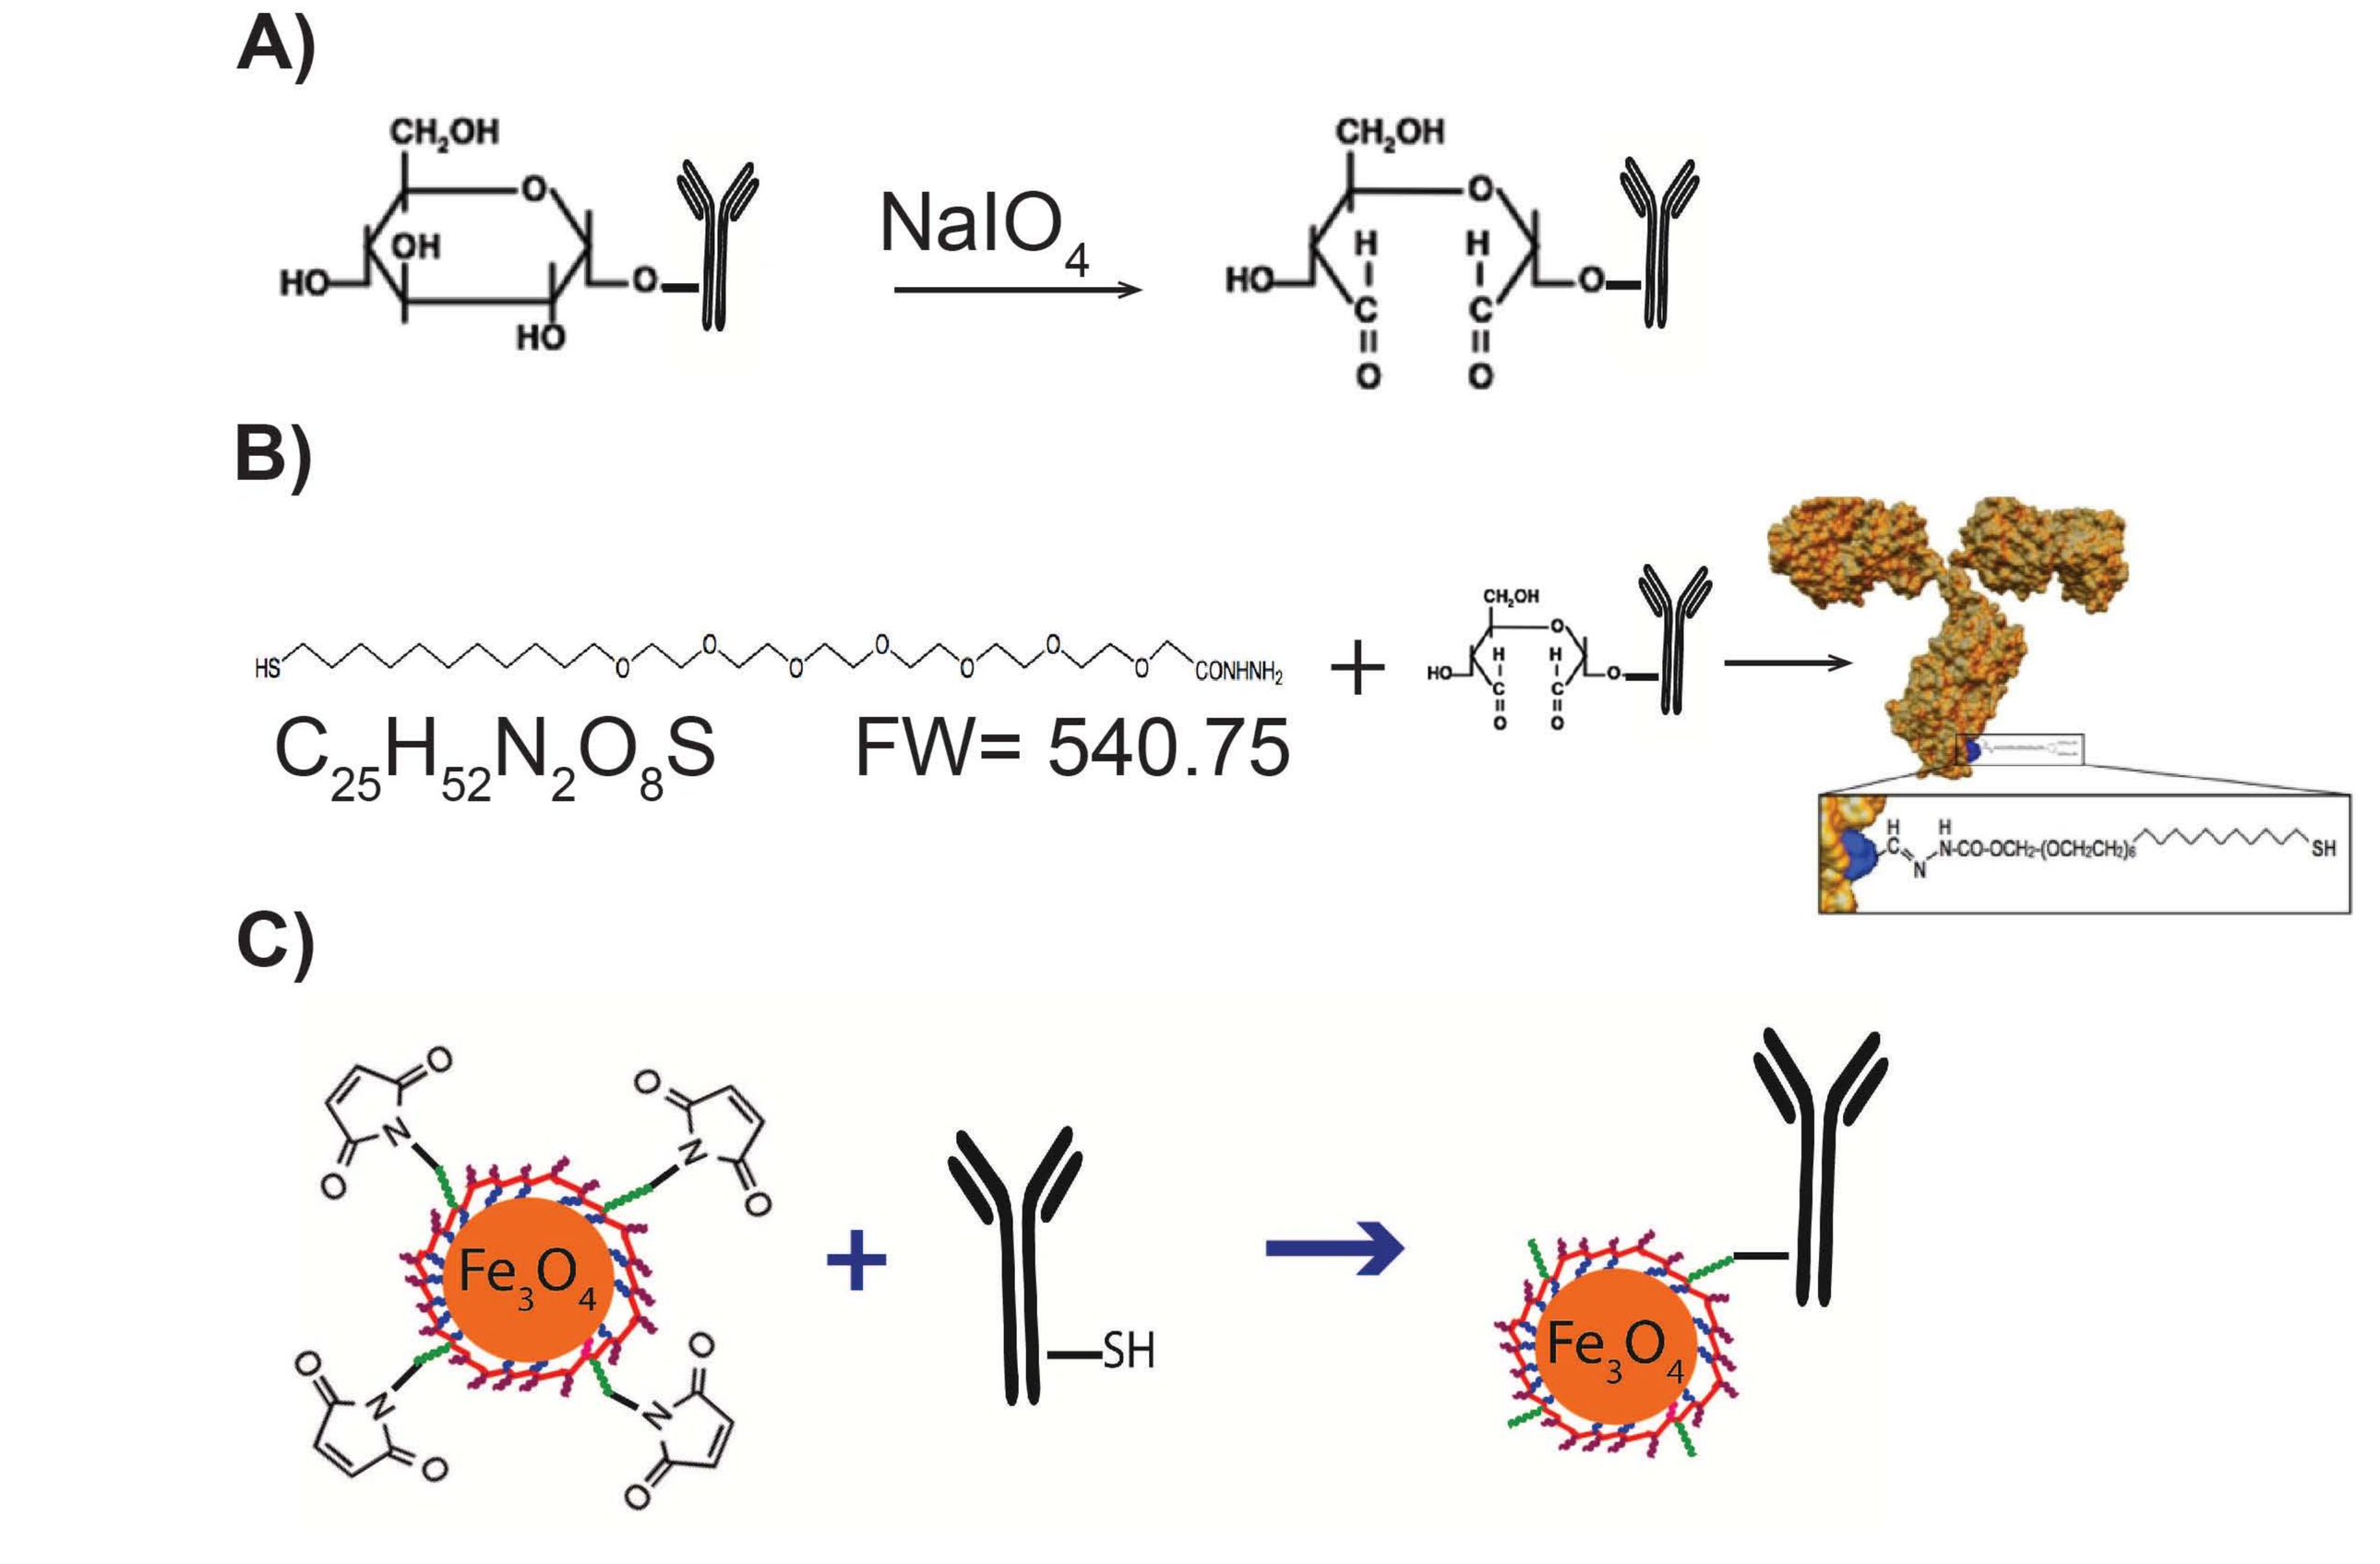
A)
NaIO4
B)
C25H52N2O8S FW= 540.75
C)
SH

Figure 4. Directional conjugation of a monoclonal antibody to the Fe_3O_4 micelle
Hydroxyl oxidation mediated by sodium periodate yielding an aldehyde group (**A**). Hydrazine - aldehyde interaction yield a hydrazone bond (**B**). The linker is a heterobifunctional molecule with a thiol opposite to the hydrazine group. By using a maleimide functionalized pegylated phospholipid to coat the surface of the Fe_3O_4 micelle an antibody can be conjugated via a maleimide-thiol bond (**C**).

An ELISA assay was conducted to estimate the number of functional antibodies conjugated to micelles (**Figure 5**). A key cancer biomarker is the epidermal growth factor receptor (EGFR), known to be highly expressed in epidermoid carcinoma A-431 cell line. Micellar anti-EGFR was estimated to have 55 ng of antibody translating into 4 antibodies per micelle (**Figure 5A**). To validate our results, micellar OC125 was generated and estimated to have 43 ng translating into 4 antibodies per micelle (**Figure 5B**). OC125 antibody is known to react with 80% of ovarian neoplasms.

Michaelis Menten

$$S = \frac{V * Km}{Vmax - V} \longrightarrow Antibody = \frac{Absorbance * Km}{Max\ absorbance - Absorbance}$$

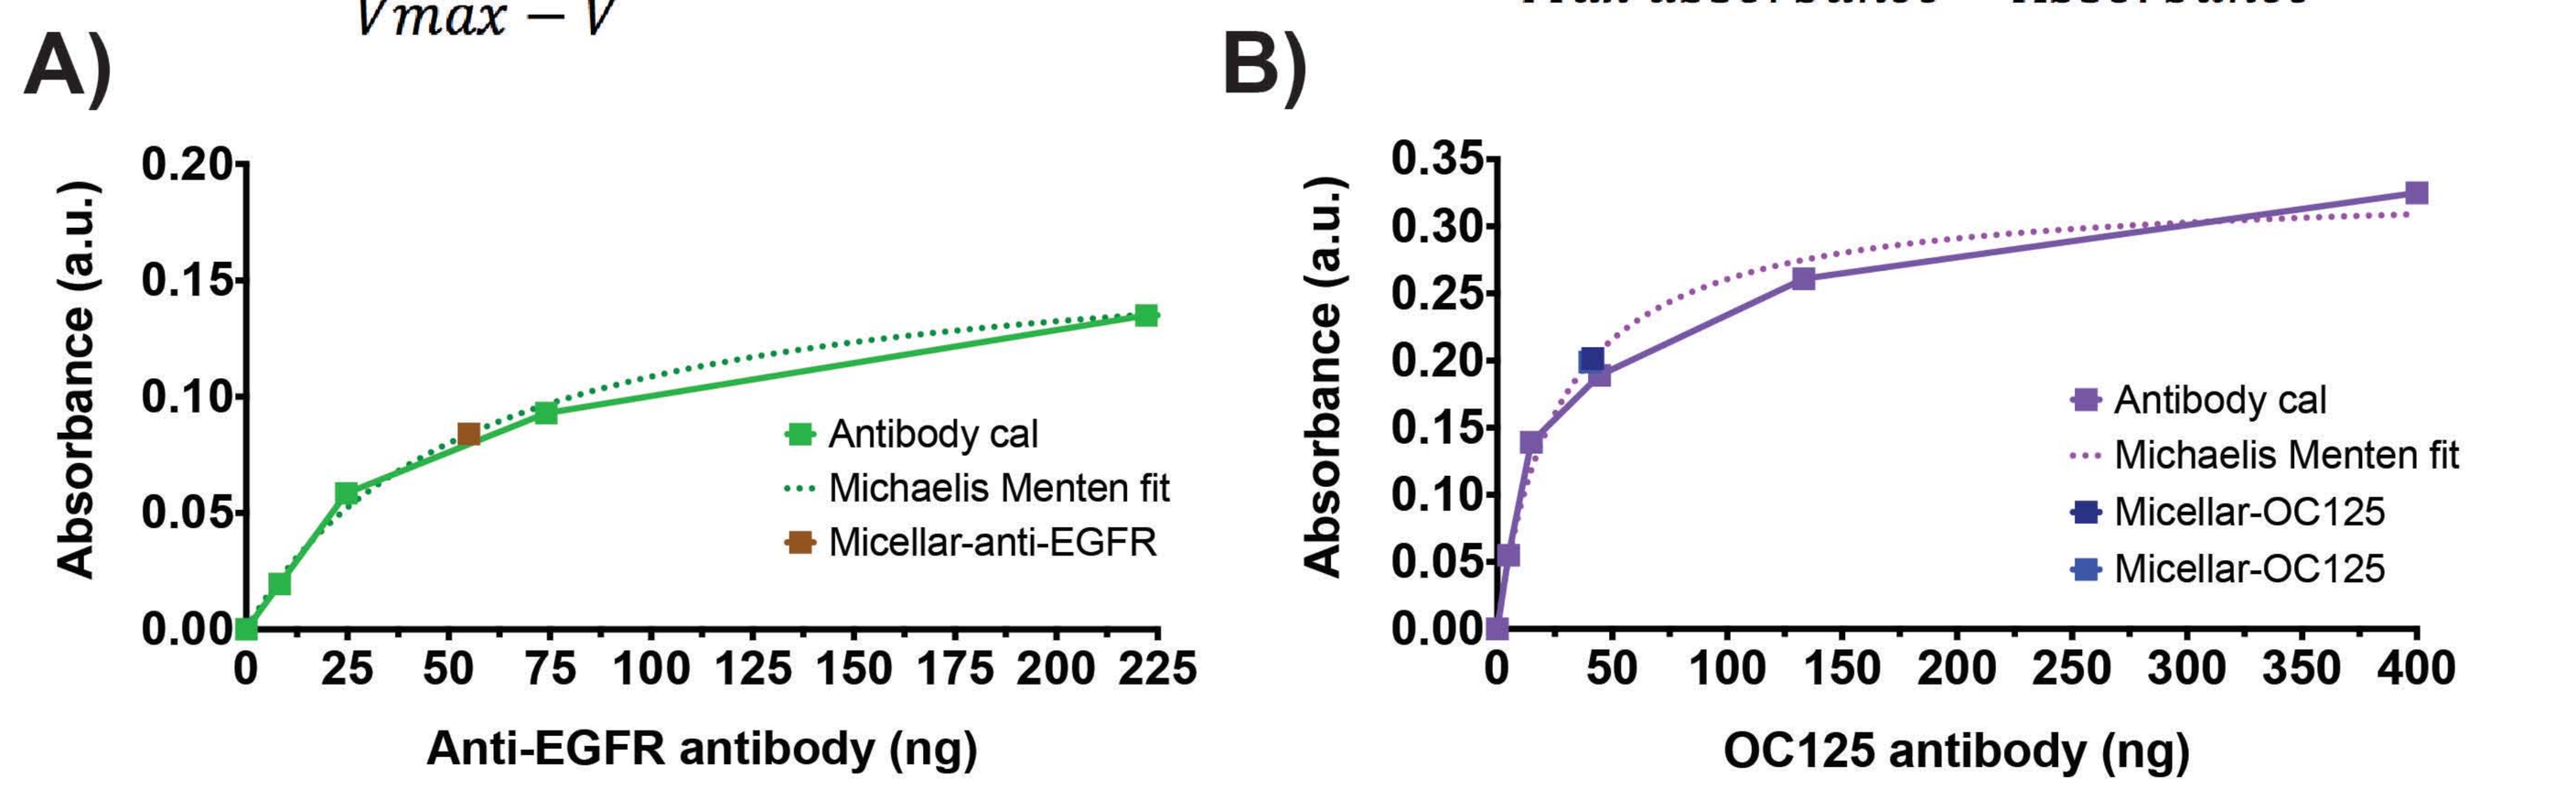
A)
Absorbance (a.u.)
Anti-EGFR antibody (ng)
Antibody cal
Michaelis Menten fit
Micellar-anti-EGFR
B)
Absorbance (a.u.)
OC125 antibody (ng)
Antibody cal
Michaelis Menten fit
Micellar-OC125
Micellar-OC125

Figure 5. Estimation of antibody conjugated to Fe_3O_4 micelles.
ELISA assay analyzed using Michaelis Menten (MM) fit[4]. Constants for the anti-EGFR antibody are Vmax = 0.17 and Km= 55.85 with R^2=0.9958 (**A**). Constants for the OC125 antibody are Vmax = 0.33 and Km= 26.41 with R^2=0.9841 (**B**).

Determination of *in vitro* molecular specificity is required prior to *in vivo* evaluation.

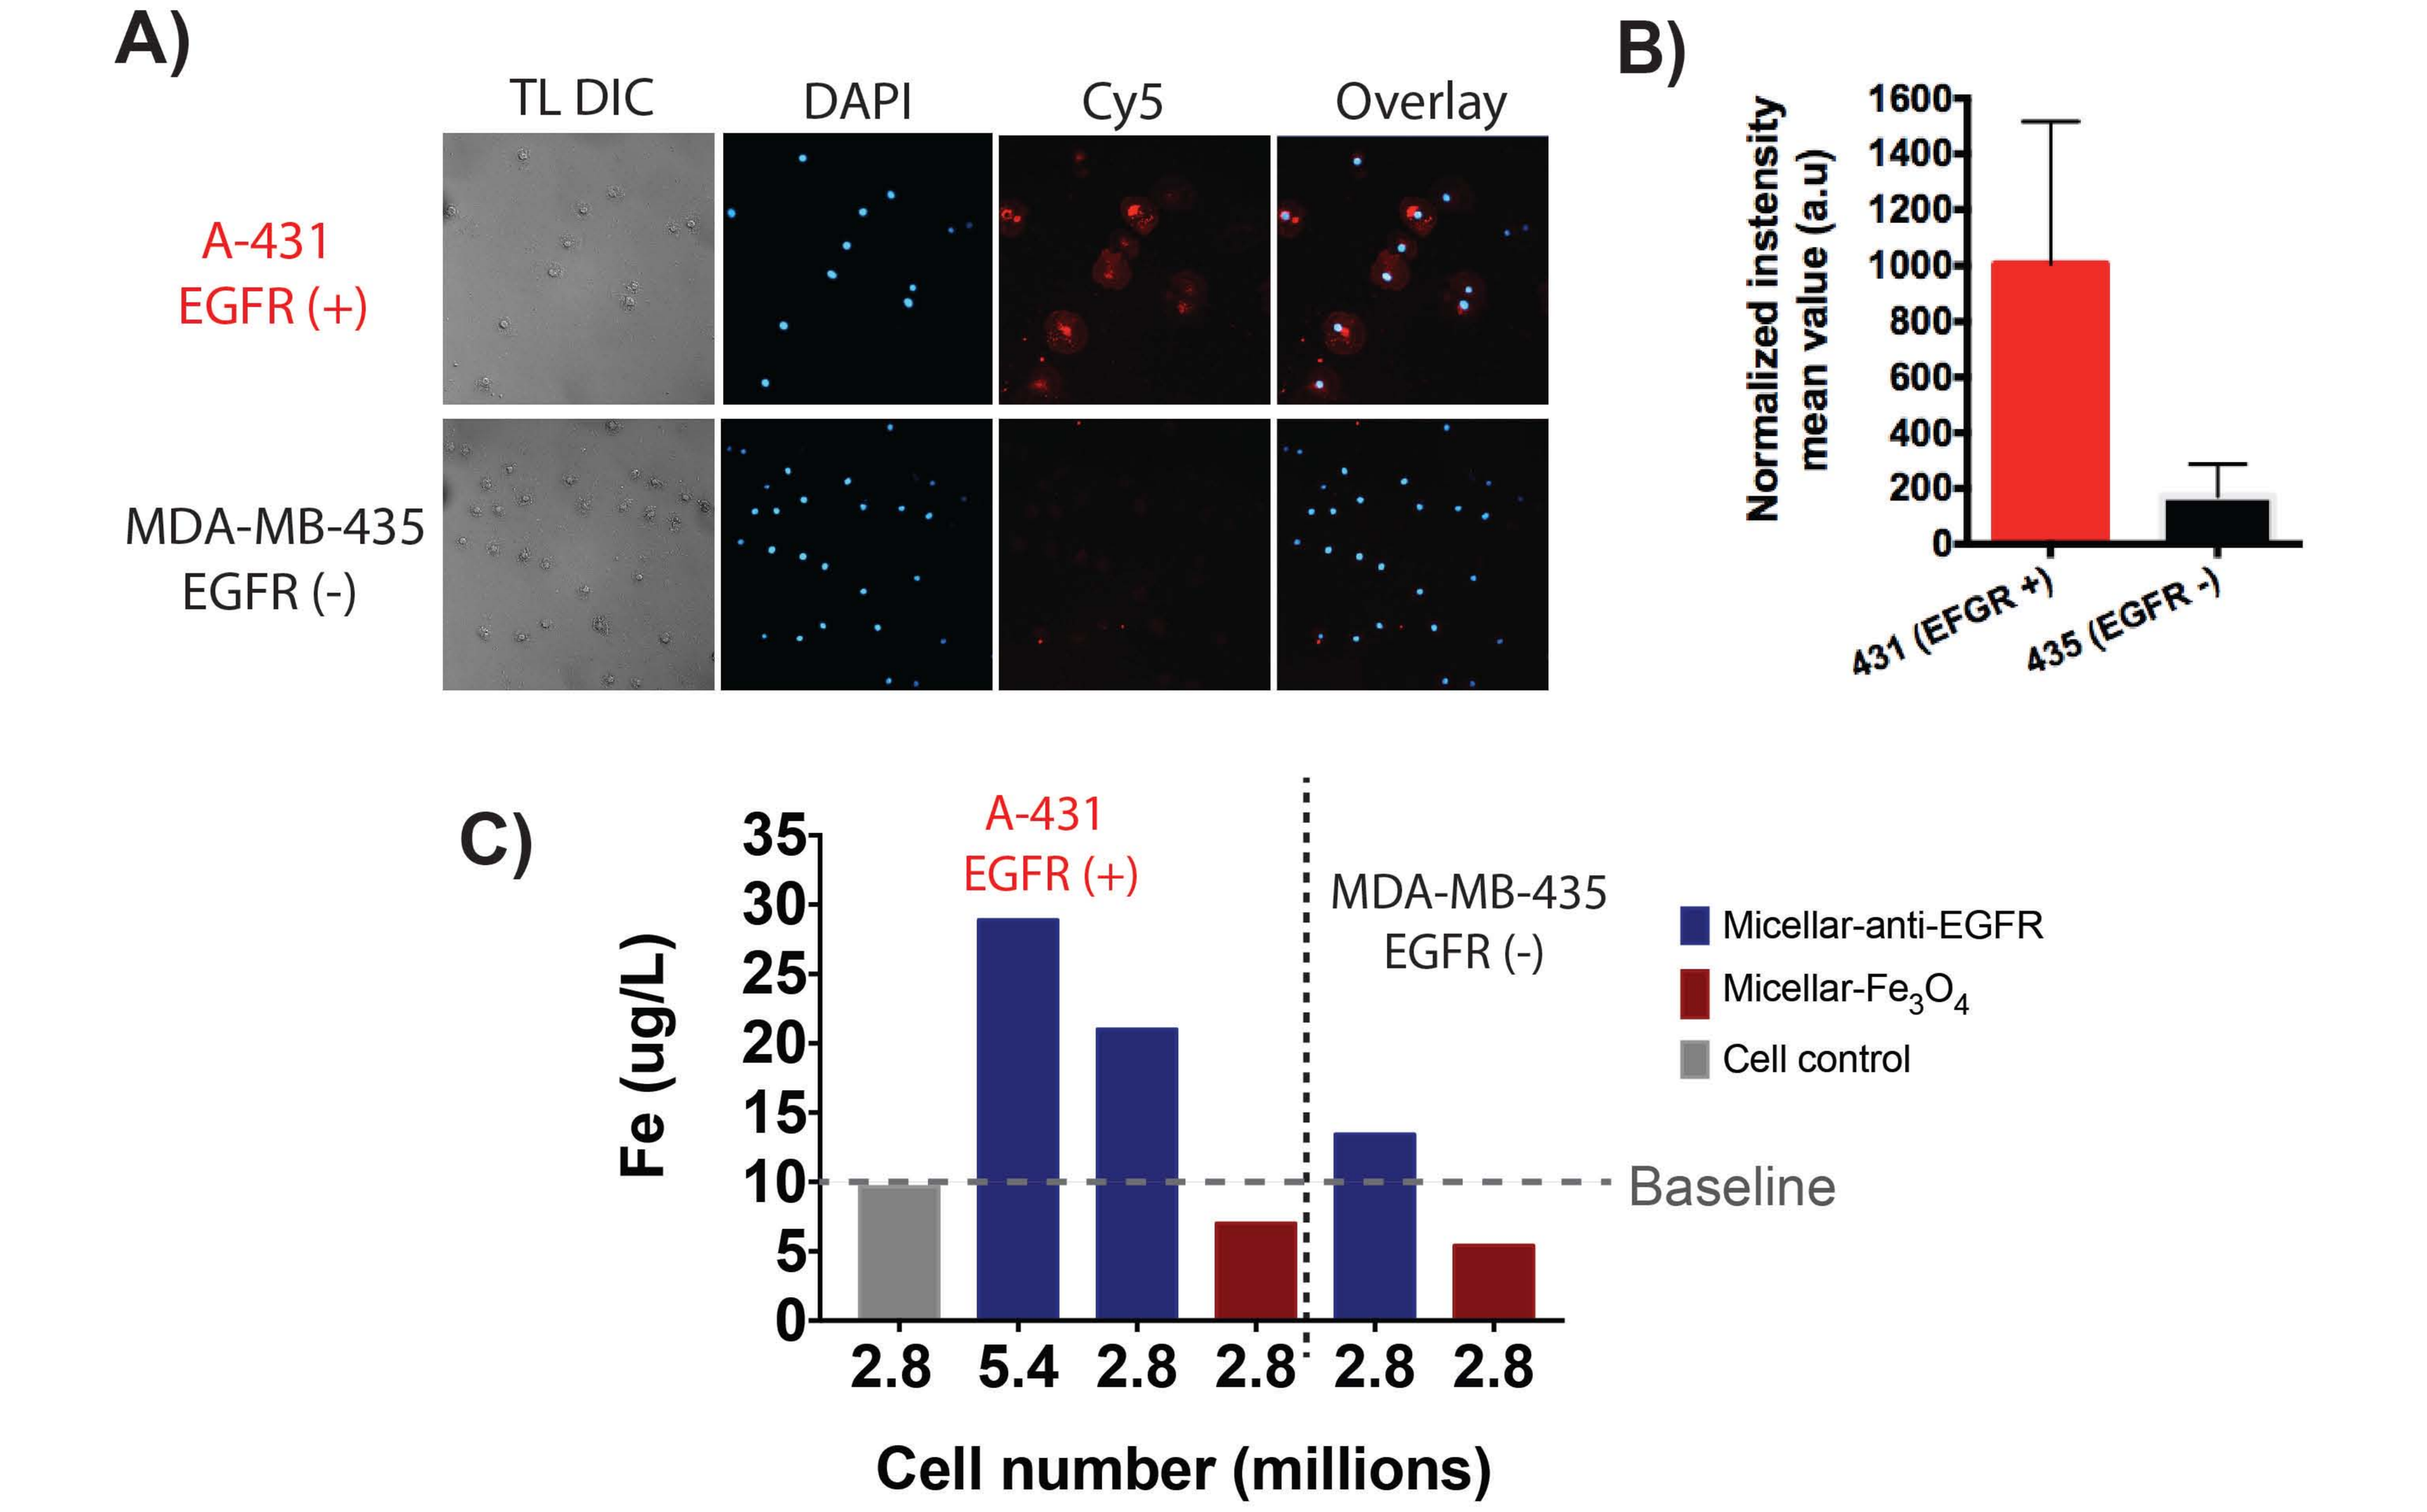
A)
TL DIC
DAPI
Cy5
Overlay
A-431
EGFR (+)
MDA-MB-435
EGFR (-)
B)
Normalized instensity mean value (a.u)
431 (EFGR +)
435 (EGFR -)
C)
Fe (ug/L)
A-431
EGFR (+)
MDA-MB-435
EGFR (-)
Micellar-anti-EGFR
Micellar-Fe3O4
Cell control
Baseline
Cell number (millions)

Figure 6. Micellar anti-EGFR exhibits molecular specificity
Fluorescence labeling with alexa 647 dye of anti-EGFR followed by micellar conjugation to interrogate molecular specific receptor-antibody interaction with EGFR expressing cell line (**A, B**). Interrogation of micellar uptake via ICP-MS on A-431 or MDA-MB-435 cell lines (**C**).

Fluorescence microscopy conducted to evaluate micellar anti-EGFR specificity, resulting in a 5-fold differential between EGFR expressing A-431 and MDA-MB-435 cell lines (**Figure 6A, B**). Uptake of micellar anti-EGFR versus its non-targeted counterpart resulted in negligible iron detection of non-targeted micelles in either cell line. EGFR targeted micelles exhibited a 3-fold increase in A-431 in comparison to the negative cell line (**Figure 6C**).

Conclusion

The development of a monodisperse, reproducible micellar Fe_3O_4 formulation, stealth towards opsonization, and displaying at least 2 hrs *in vivo* circulation has been presented. Directional antibody conjugation resulted in an estimation of 4 antibodies per micelle. Antibody functionality has been demonstrated with two different systems. Molecular specificity determined by fluorescence microscopy and intracellular iron detection mediated by receptor-antibody interactions.

Future work includes *in vitro* and *in vivo* evaluation of targeted micellar Fe_3O_4 to push the boundaries of sensitive cancer detection using magnetic relaxometry.

References

1) Vreeland, EC, et al., *Chem Mater.*, 27: 6059-6066, 2015.
2) Wiekhorst F, et al., *Pharm Res.* 29: 1189-1202, 2012.
3) Kumar S, et al., *Nature Protocols.* v. 3 (2): 314-320, 2008.
4) Vira S, et al. *Anal Biochem.* 402 (2): 146-150, 2010.